Short-Term Efficacy and Safety of Abrocitinib by Baseline Disease Severity in Patients With Moderate-to-Severe Atopic Dermatitis

Zakiya P. Rice,[1] Melinda J. Gooderham,[2] Mark G. Lebwohl,[3] Eric L. Simpson,[4] Mark Boguniewicz,[5] Andreas Wollenberg,[6] Irina Lazariciu,[7] Gary L. Chan,[8] Justine Alderfer,[9] Melissa Watkins[10]

[1]Dermatology Associates of Georgia, Atlanta, GA, USA; [2]SKiN Centre for Dermatology, Queen's University and Probity Medical Research, Peterborough, ON, Canada; [3]Department of Dermatology, Icahn School of Medicine at Mount Sinai, New York, NY, USA; [4]Department of Dermatology, Oregon Health & Science University, Portland, OR, USA; [5]Department of Pediatrics, National Jewish Health and University of Colorado School of Medicine, Denver, CO, USA; [6]Ludwig Maximilian University, Munich, Germany; [7]Department of Biostatistics, IQVIA, QC, Canada; [8]Pfizer Inc., Groton, CT, USA; [9]Pfizer Inc., Collegeville, PA, USA; [10]Pfizer Inc., New York, NY, USA

BACKGROUND

- Atopic dermatitis (AD) is a chronic inflammatory skin disease associated with substantial patient burden that increases with greater disease severity[1,2]
 - Despite treatment with systemic therapies, patients with moderate or severe AD often report significantly worse outcomes than patients with mild AD, including severe itch, pain, and greater impact on quality of life
- There is a need for more effective therapies in patients with moderate or severe disease
- Abrocitinib is an oral, once-daily, Janus kinase 1–selective inhibitor approved for the treatment of adults and adolescents with moderate-to-severe AD[3-5]
- Abrocitinib was efficacious and well tolerated in patients when administered as monotherapy or in combination with background topical therapy in multiple phase 3 clinical trials[6-8]

OBJECTIVE

- To evaluate the efficacy and safety of abrocitinib in patients with moderate-to-severe AD classified by baseline disease severity

METHODS

Study Design and Assessments

- Data were analyzed post hoc from clinical trials with abrocitinib administered as monotherapy (pooled phase 2b [NCT02780167] and phase 3 JADE MONO-1 [NCT03349060] and JADE MONO-2 [NCT03575871]) or in combination with topical therapy (JADE COMPARE; NCT03720470)
- The study designs and assessments are shown in **Figure 1**

Figure 1. Study Designs and Assessments

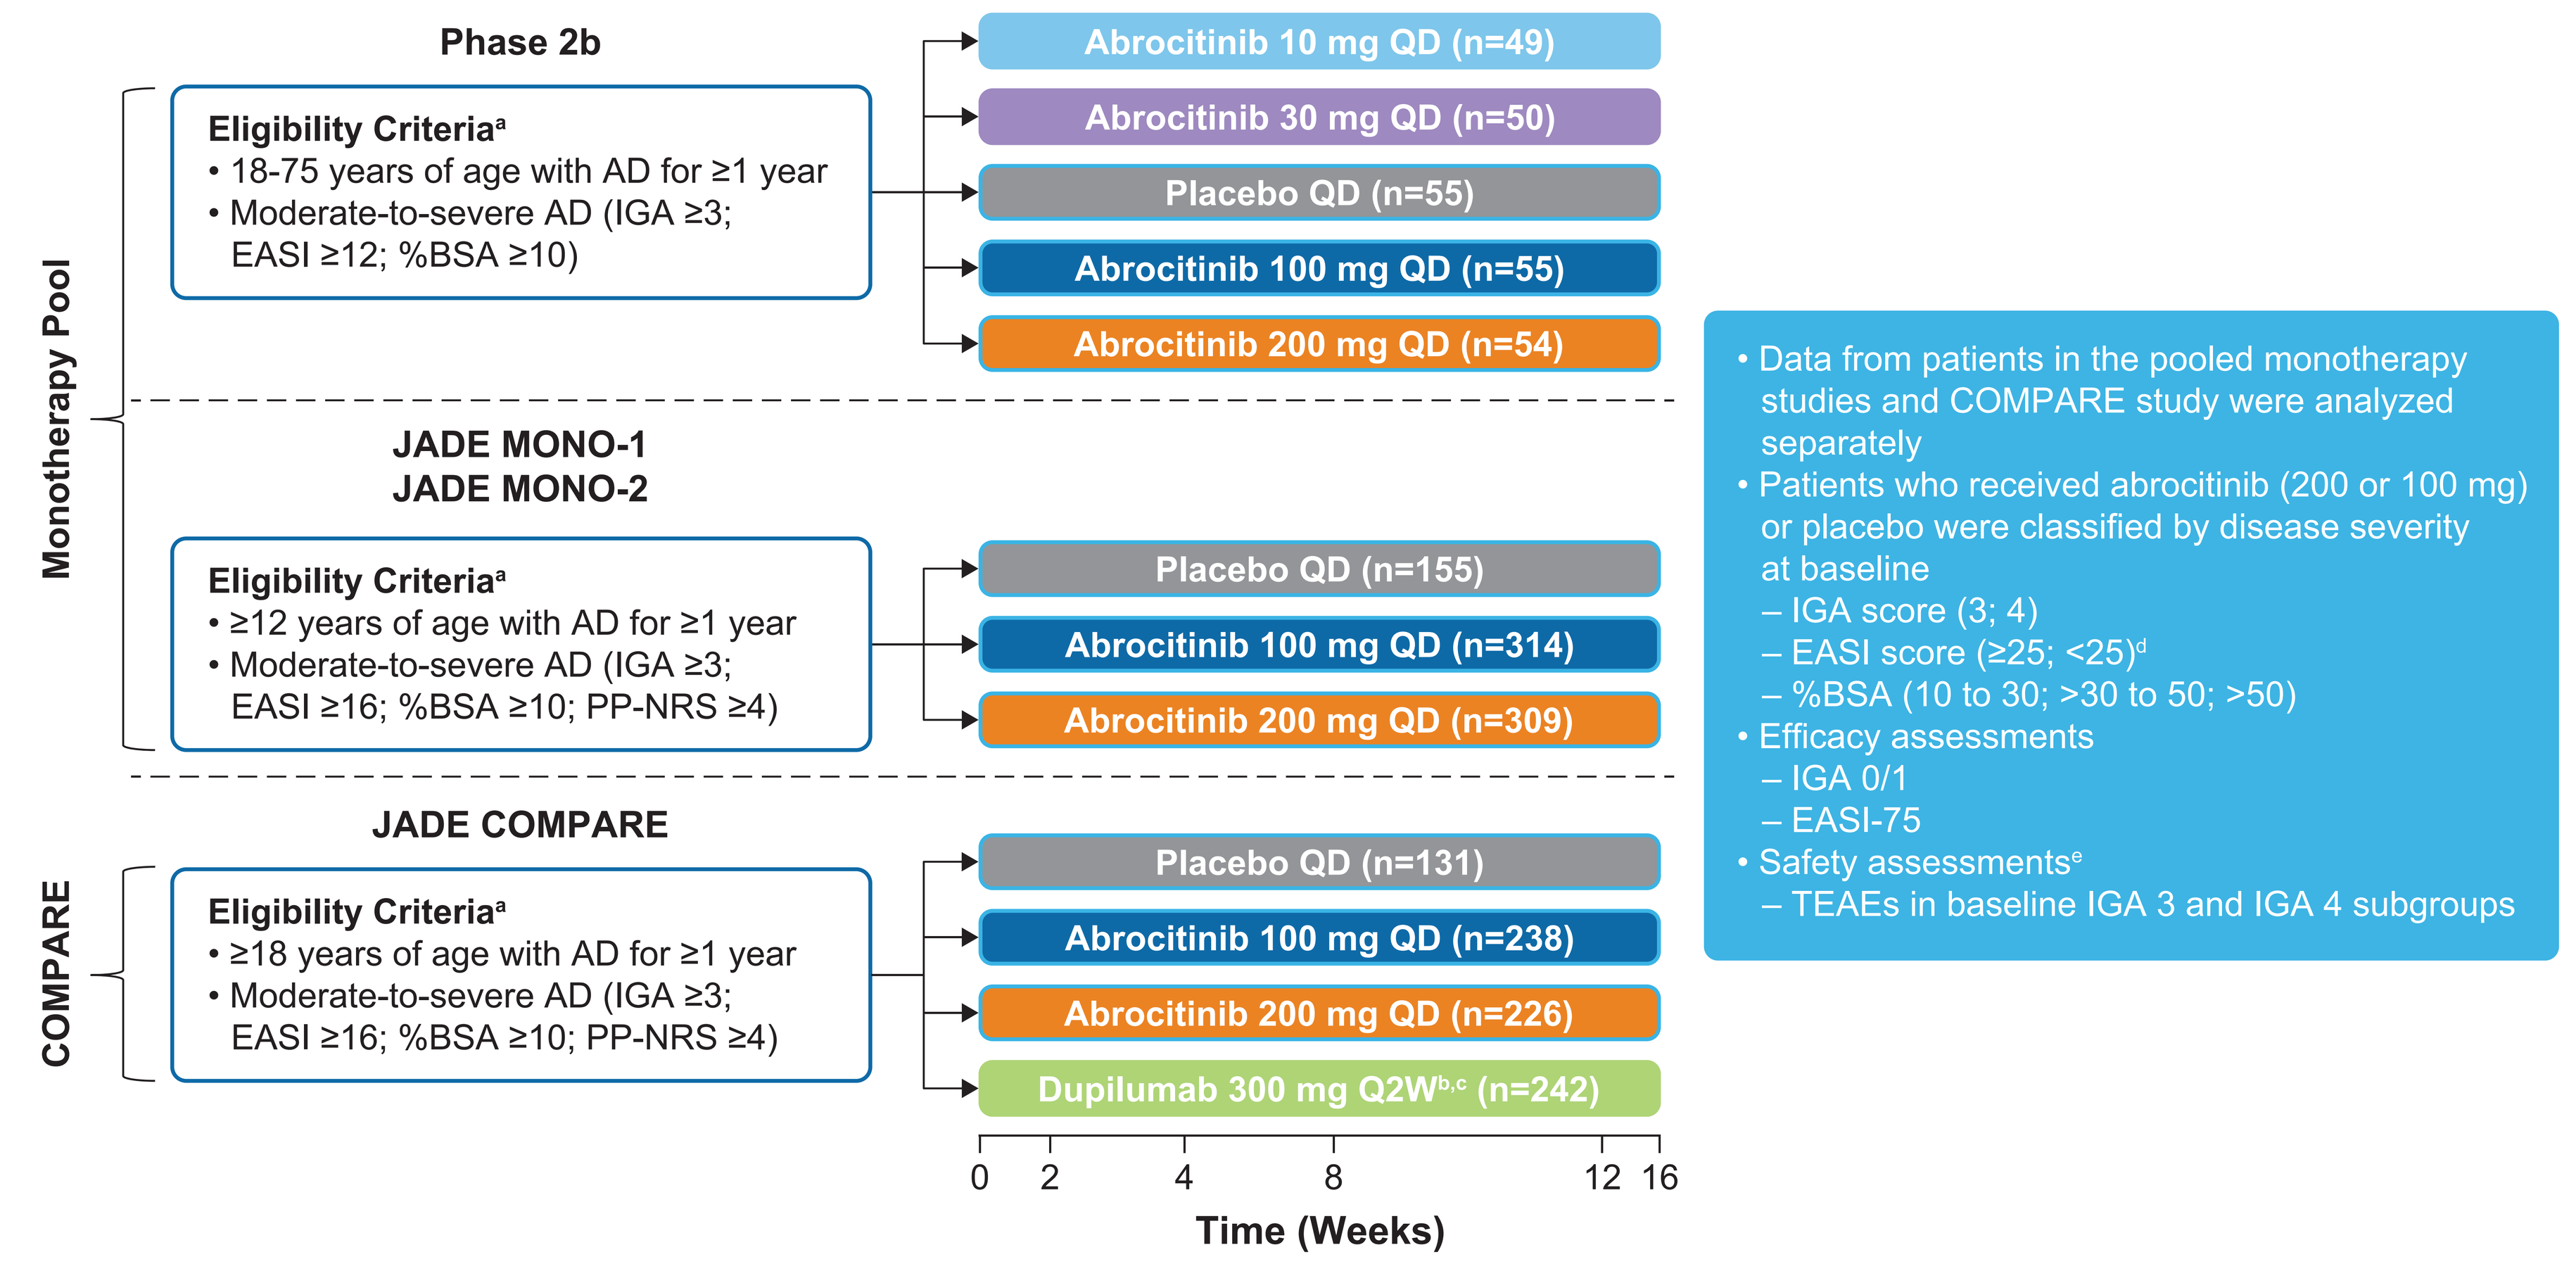
Phase 2b
Eligibility Criteria[a]
• 18-75 years of age with AD for ≥1 year
• Moderate-to-severe AD (IGA ≥3; EASI ≥12; %BSA ≥10)
Abrocitinib 10 mg QD (n=49)
Abrocitinib 30 mg QD (n=50)
Placebo QD (n=55)
Abrocitinib 100 mg QD (n=55)
Abrocitinib 200 mg QD (n=54)
Monotherapy Pool
JADE MONO-1
JADE MONO-2
Eligibility Criteria[a]
• ≥12 years of age with AD for ≥1 year
• Moderate-to-severe AD (IGA ≥3; EASI ≥16; %BSA ≥10; PP-NRS ≥4)
Placebo QD (n=155)
Abrocitinib 100 mg QD (n=314)
Abrocitinib 200 mg QD (n=309)
JADE COMPARE
COMPARE
Eligibility Criteria[a]
• ≥18 years of age with AD for ≥1 year
• Moderate-to-severe AD (IGA ≥3; EASI ≥16; %BSA ≥10; PP-NRS ≥4)
Placebo QD (n=131)
Abrocitinib 100 mg QD (n=238)
Abrocitinib 200 mg QD (n=226)
Dupilumab 300 mg Q2W[b,c] (n=242)
0 2 4 8 12 16
Time (Weeks)
• Data from patients in the pooled monotherapy studies and COMPARE study were analyzed separately
• Patients who received abrocitinib (200 or 100 mg) or placebo were classified by disease severity at baseline
– IGA score (3; 4)
– EASI score (≥25; <25)[d]
– %BSA (10 to 30; >30 to 50; >50)
• Efficacy assessments
– IGA 0/1
– EASI-75
• Safety assessments[e]
– TEAEs in baseline IGA 3 and IGA 4 subgroups

%BSA, percentage of body surface area; AD, atopic dermatitis; EASI, Eczema Area and Surface Index; EASI-75, ≥75% improvement on the EASI; IGA, Investigator's Global Assessment; IGA 0/1, IGA score of 0 (clear) or 1 (almost clear) with a ≥2-point improvement from baseline; IGA 3, IGA score of 3 (moderate); IGA 4, IGA score of 4 (severe); PP-NRS, Peak Pruritus Numerical Rating Scale (used with permission from Regeneron Pharmaceuticals, Inc., and Sanofi); Q2W, once every 2 weeks; QD, once daily; TEAE, treatment-emergent adverse event.

[a]Inadequate response or intolerance to topical medication or requirement for systemic therapy to control AD.
[b]Active control arm; data not included in this analysis.
[c]After a 600-mg loading dose of subcutaneous duplilumab.
[d]The mean baseline EASI score for the monotherapy pool was 25.
[e]The safety analysis also included patients from the JADE REGIMEN (NCT03627767; open-label phase) and JADE EXTEND (NCT03422822; April 2020 data cut) studies.

RESULTS

Efficacy

- In both the JADE monotherapy and the JADE COMPARE populations, a greater proportion of patients achieved IGA 0/1 or EASI-75 responses with abrocitinib 200 mg and abrocitinib 100 mg compared with placebo at week 12 across all subgroups of disease severity as classified by baseline IGA or EASI scores (**Figure 2**)

Figure 2. Efficacy at Week 12 in Baseline Disease Severity Subgroups by IGA and EASI Scores

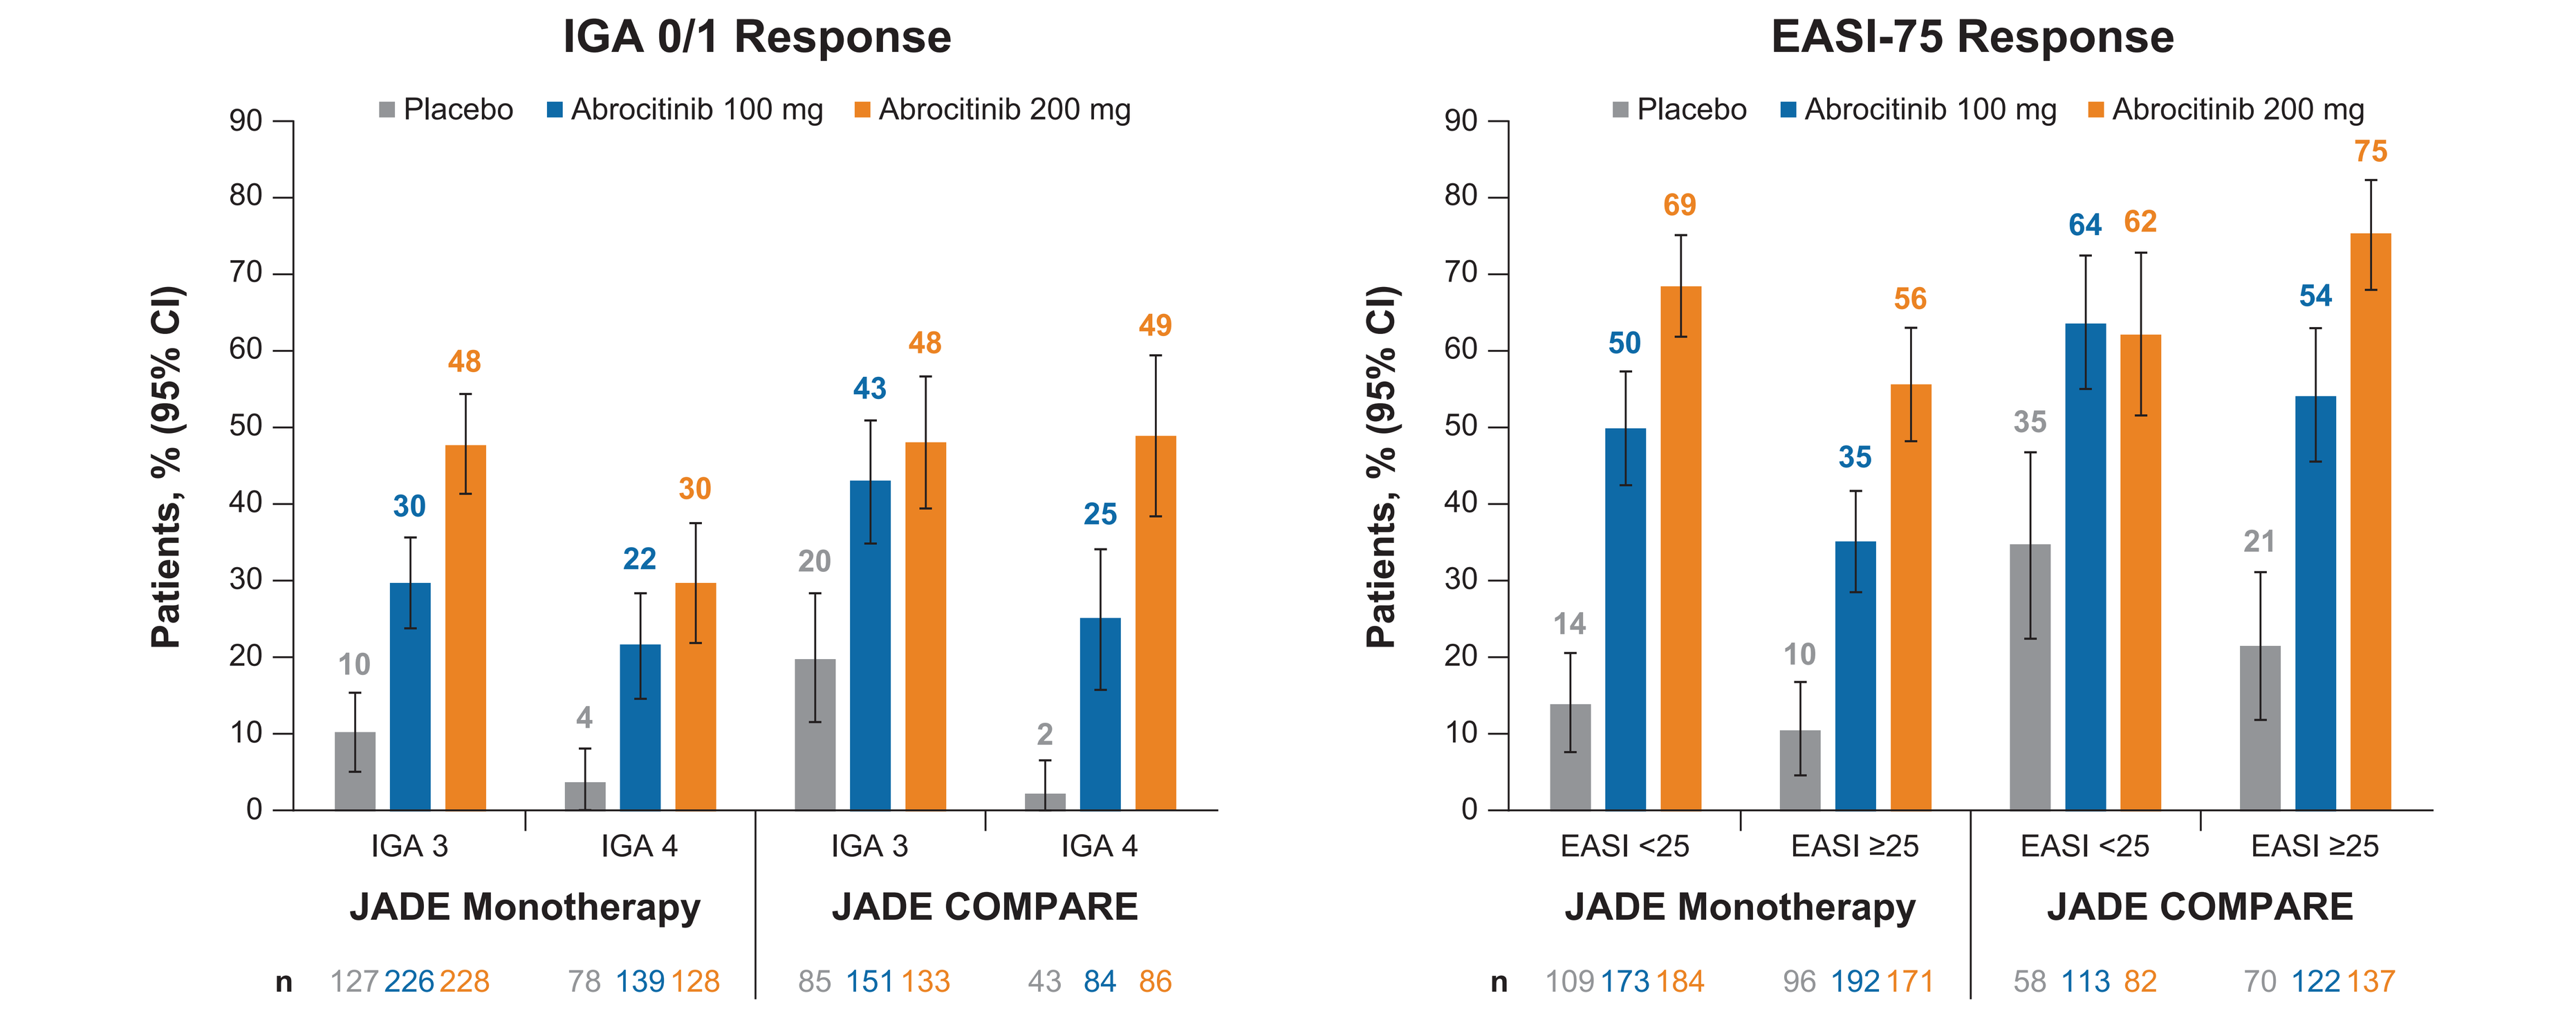
IGA 0/1 Response
Placebo ■ Abrocitinib 100 mg ■ Abrocitinib 200 mg
Patients, % (95% CI)
IGA 3: 10 30 48
IGA 4: 4 22 30
IGA 3: 20 43 48
IGA 4: 2 25 49
JADE Monotherapy
JADE COMPARE
n 127 226 228 | 78 139 128 | 85 151 133 | 43 84 86
EASI-75 Response
EASI <25: 14 50 69
EASI ≥25: 10 35 56
EASI <25: 35 64 62
EASI ≥25: 21 54 75
JADE Monotherapy
JADE COMPARE
n 109 173 184 | 96 192 171 | 58 113 82 | 70 122 137

IGA, Investigator's Global Assessment; IGA 0/1, IGA score of 0 (clear) or 1 (almost clear) with a ≥2-point improvement from baseline; IGA 3, IGA score of 3 (moderate); IGA 4, IGA score of 4 (severe).

- In the subgroups classified by %BSA (percentage of body surface area), a greater proportion of patients achieved IGA 0/1 response with abrocitinib 200 mg and abrocitinib 100 mg than placebo at week 12 (**Figure 3**)
- In the JADE monotherapy population, efficacy responses occurred in a dose-dependent manner across all subgroups of baseline disease severity (**Figures 2** and **3**)

Figure 3. Efficacy at Week 12 in Baseline Disease Severity Subgroups by %BSA

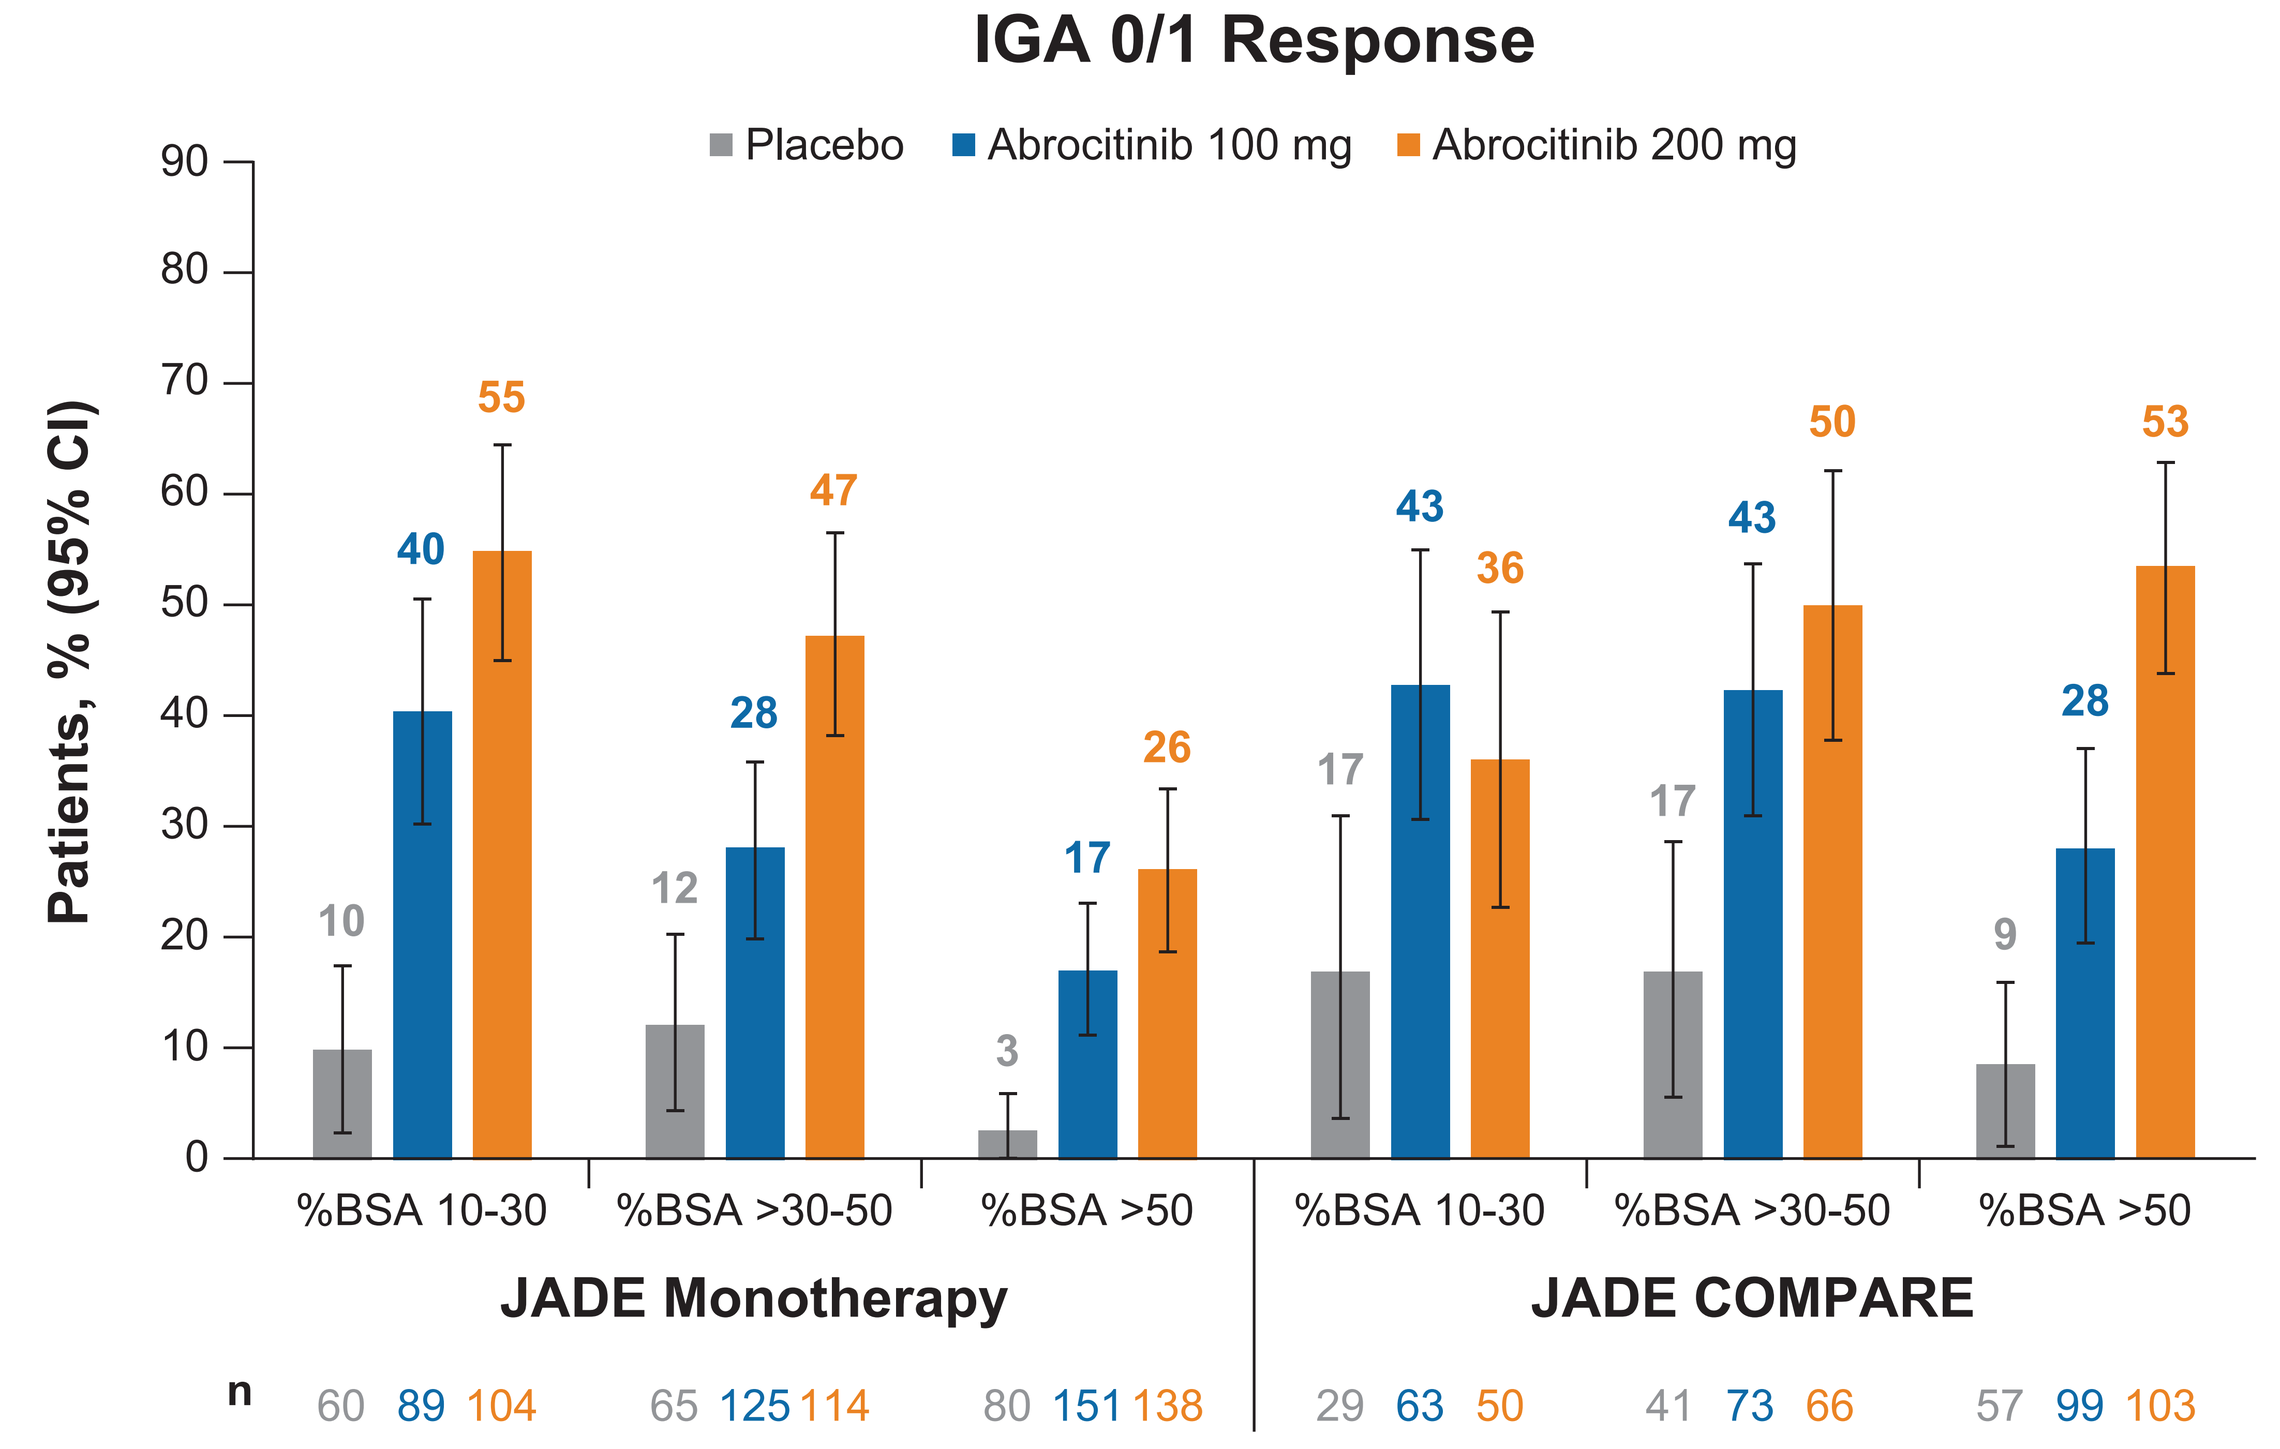
IGA 0/1 Response
Placebo ■ Abrocitinib 100 mg ■ Abrocitinib 200 mg
Patients, % (95% CI)
%BSA 10-30: 10 40 55
%BSA >30-50: 12 28 47
%BSA >50: 3 17 26
%BSA 10-30: 17 43 36
%BSA >30-50: 17 43 50
%BSA >50: 9 28 53
JADE Monotherapy
JADE COMPARE
n 60 89 104 | 65 125 114 | 80 151 138 | 29 63 50 | 41 73 66 | 57 99 103

%BSA, percentage of body surface area; IGA 0/1, Investigator's Global Assessment score of 0 (clear) or 1 (almost clear) with a ≥2-point improvement from baseline.

Safety

- Overall treatment-emergent adverse events (AEs) were similar in both the baseline IGA 3 and IGA 4 subgroups
 - The rates of serious AEs, severe AEs, and AEs leading to discontinuation were also similar
- The incidence rates for serious infections were similar in both subgroups and had widely overlapping confidence intervals (**Table 1**)
- The incidence rates for all herpes zoster infections was higher in the IGA 4 subgroup than the IGA 3 subgroup (**Table 1**)
- No meaningful differences were seen in laboratory values of interest between the 2 subgroups

Table 1. Safety in Baseline Disease Severity Subgroups by IGA Score

AEs of Special Interest (per 100 PY)	IGA 3 n=1753	IGA 4 n=1103
Serious infections	2.18	2.88
Herpes zoster infections[a]	2.49	4.93

AE, adverse event; PY, person-years.
[a]The hazard ratio for IGA=4 over IGA=3 obtained in a multivariate analysis using a Cox regression model was significantly greater than 1.

CONCLUSIONS

- Abrocitinib, used as monotherapy or in combination with topical therapy, provided clinically meaningful improvements in patients with AD across various subgroups of baseline disease severity
- A greater proportion of patients achieved IGA 0/1 or EASI-75 with abrocitinib versus placebo at week 12 across all subgroups of disease severity classified by baseline IGA score, baseline EASI score, or %BSA at baseline
- Efficacy responses were dose dependent across all subgroups of baseline disease severity in the pooled monotherapy population
- AEs were similar in both the IGA 3 and IGA 4 baseline disease severity subgroups, with no unexpected safety signals

REFERENCES

1. Simpson et al. *J Am Acad Dermatol*. 2016;74:491-498.
2. Simpson et al. *JAMA Dermatol*. 2018;154:903-912.
3. Cibinqo (abrocitinib). Prescribing information. Pfizer Ltd.; September 2021.
4. Cibinqo (abrocitinib). Summary of Product Characteristics. Pfizer Europe; October 2022.
5. Cibinqo (abrocitinib). Prescribing information. Pfizer Labs; January 2022.
6. Simpson EL et al. *Lancet*. 2020;396:255-266.
7. Silverberg JI et al. *JAMA Dermatol*. 2020;156:863-873.
8. Bieber T et al. *N Engl J Med*. 2021;384:1101-1112.

ACKNOWLEDGMENTS

Editorial/medical writing support under the guidance of authors was provided by Amanda Mabhula, PhD, and Renata Cunha, PharmD, at ApotheCom, San Francisco, CA, USA, and was funded by Pfizer Inc., New York, NY, USA, in accordance with Good Publication Practice (GPP 2022) guidelines (*Ann Intern Med*. 2022; 10.7326/M22-1460). This analysis was sponsored by Pfizer Inc.

Previously presented at the International Society of Atopic Dermatitis (ISAD); October 17-19, 2022; Montréal, Québec, Canada

DISCLOSURES

ZPR is an advisory board member for Pfizer Inc., Biotech, Cassiopeia, and Medscape; a consultant for Pfizer Inc., Brickell Biotech, Cassiopeia, Dermira, and Regeneron Pharmaceuticals/Sanofi; has received funding from Anacor Pharmaceuticals, Celgene, Galderma, Regeneron/Sanofi Genzyme, Merck & Co., and AbbVie; and has been a speaker for Pfizer Inc., Dermira, the International Hyperhidrosis Society, PRIME, and Regeneron Pharmaceuticals/Sanofi. **MJG** has received grants, personal fees, honoraria, and/or nonfinancial support from Pfizer Inc., AbbVie, Amgen, AkrosPharma, Arcutis, Bristol Myers Squibb, Boehringer Ingelheim, Celgene, Dermira, Eli Lilly and Company, Galderma, Janssen, Kyowa Kirin, LEO Pharma, MedImmune, Merck, Novartis, Roche, Sanofi Genzyme, Regeneron, Sun Pharma, UCB, Bausch Health (Valeant), Aslan, Dermavant and Meiji. **MGL** is an employee of Mount Sinai and receives research funds from Abbvie, Amgen, Arcutis, Avotres, Boehringer Ingelheim, Cara Therapeutics, Dermavant Sciences, Eli Lilly and Company, Incyte, Janssen Research & Development, LLC, Ortho Dermatologics, Regeneron, and UCB, Inc. and is a consultant for Pfizer Inc., Aditum Bio, Almirall, AltruBio Inc., AnaptysBio, Arcutis, Inc., Arena Pharmaceuticals, Aristea Therapeutics, Arrive Technologies, Avotres Therapeutics, BiomX, Brickell Biotech, Boehringer Ingelheim, Bristol Myers Squibb, Cara Therapeutics, Castle Biosciences, Corevitas, Dermavant Sciences, Dr. Reddy's Laboratories, Evelo Biosciences, Evommune, Inc., Facilitation of International Dermatology Education, Forte Biosciences, Foundation for Research and Education in Dermatology, Helsinn Therapeutics, Hexima Ltd., LEO Pharma, Meiji Seika Pharma, Mindera, Seanergy, and Verrica. **ELS** has received grants from Pfizer Inc., Eli Lilly and Company, Kyowa Kirin, LEO Pharma, Merck, and Regeneron and personal fees from Pfizer Inc., Bausch Health (Valeant), Dermira, Eli Lilly and Company, Galderma, LEO Pharma, Menlo Therapeutics, Novartis, Regeneron, and Sanofi-Genzyme. **MB** has been an investigator for Regeneron and Incyte and an advisor for Pfizer Inc., AbbVie, Eli Lilly and Company, Incyte, Janssen, LEO Pharma, Regeneron, and Sanofi Genzyme. **AW** has been an advisor, speaker, or investigator for Pfizer Inc., Almirall, Beiersdorf, Bioderma, Chugai, Galapagos, Galderma, Hans Karrer, LEO Pharma, Eli Lilly and Company, L'Oreal, Maruho, MedImmune, Novartis, Pierre Fabre, Regeneron, Santen, and Sanofi. **IL** is an employee of IQVIA, who were paid contractors to Pfizer Inc. in the development of this poster and in providing statistical support. **GLC**, **JA**, and **MW** are employees and shareholders of Pfizer Inc.

Copies of this poster obtained through this QR code, are for your personal use only and may not be reproduced without permission from the authors.

Copyright © 2023. All rights reserved.

Presented at the Winter Clinical Dermatology Conference; January 13-18, 2023; Kohala Coast, Hawaii