Long-Term Efficacy of Abrocitinib in Adolescents and Adults With Moderate-to-Severe Atopic Dermatitis: A Post Hoc Analysis of JADE EXTEND

Amy S. Paller,[1] Michael J. Cork,[2] Carsten Flohr,[3] Christine Bangert,[4] Stephan Weidinger,[5] John Nesnas,[6] Saleem A. Farooqui,[7] Pinaki Biswas,[8] Melissa Watkins,[8] Herwig Koppensteiner[9]

[1]Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [2]Sheffield Dermatology Research, IICD, University of Sheffield and Sheffield Children's Hospital, Sheffield, United Kingdom; [3]St John's Institute of Dermatology, Guy's and St Thomas' NHS Foundation Trust, King's College London, London, United Kingdom; [4]Medical University of Vienna, Vienna, Austria; [5]University Hospital Schleswig-Holstein, Kiel, Germany; [6]Pfizer Ltd., Tadworth, Surrey, United Kingdom; [7]Pfizer R & D UK Ltd., Sandwich, Kent, United Kingdom; [8]Pfizer Inc., New York, NY, USA; [9]Pfizer Corporation Austria GmbH, Vienna, Austria

INTRODUCTION

- Abrocitinib is an oral, once-daily, Janus kinase 1–selective inhibitor approved for the treatment of moderate-to-severe atopic dermatitis (AD) in adolescents and adults in several countries, including Great Britain and the United States[1,2]
- Pivotal phase 3 trials have demonstrated short-term efficacy of abrocitinib over 12 or 16 weeks in adolescent and adult patients with moderate-to-severe AD[3-6]

OBJECTIVE

- To assess abrocitinib treatment efficacy up to 96 weeks in adolescents and adults with moderate-to-severe AD who enrolled in the ongoing phase 3 extension trial JADE EXTEND (NCT03422822)

METHODS

- This interim analysis included adolescent (aged 12 to <18 years) and adult (aged ≥18 years) patients from qualifying parent phase 3 JADE trials who subsequently enrolled in JADE EXTEND (data cut-off date: September 25, 2021; **Supplementary Figure S1**; accessed via the QR code)
- Additional information on the study design, treatment, assessments and statistical analysis are described in the **Supplementary Information**

RESULTS

Patients

- This analysis included 357 adolescent and 1309 adult patients
- Baseline disease characteristics were largely comparable across both patient groups (**Supplementary Table S1**)
 - Adolescent patients tended to have more severe disease than adult patients (Investigator's Global Assessment [IGA] score of 4: 42.3% vs 36.7%) and greater median percentage body surface area affected by AD (48.0% [95% CI, 33.0-68.1] vs 43.0% [29.0-62.9]) at baseline

Efficacy Responses Up to Week 96 of Abrocitinib Treatment in Adolescent and Adult Patients

- In both adolescents and adults and regardless of abrocitinib dose, substantial proportions of patients achieved clinically meaningful responses and improvements in clinical and patient-reported efficacy measures, including high-threshold efficacy endpoints, up to week 96 of treatment (**Figure 1** and **Supplementary Figure S2**)
 - Dose-dependent efficacy was observed for both patient groups; treatment with abrocitinib 200 mg once daily (QD) resulted in numerically higher proportions of responders than abrocitinib 100 mg QD
- At week 96, the proportions of responders were largely comparable for both doses of abrocitinib in both adolescents and adults

CONCLUSIONS

- This analysis demonstrates the short-term and long-term efficacy of treatment with abrocitinib 100 mg and abrocitinib 200 mg up to 96 weeks for both adolescent and adult patients with moderate-to-severe AD
- Substantial proportions of adolescents and adults achieved high-threshold efficacy endpoints over the short-term and long-term with either dose of abrocitinib
- Dose-dependent efficacy was observed for both adolescents and adults over the short-term and long-term, including for high-threshold efficacy endpoints

Figure 1. Proportions of Adolescent and Adult Patients Who Achieved (A) EASI-75, (B) EASI-90, (C) EASI-100, (D) PP-NRS4, (E) PP-NRS 0/1, and (F) IGA 0/1 Responses Over 96 Weeks of Abrocitinib Treatment

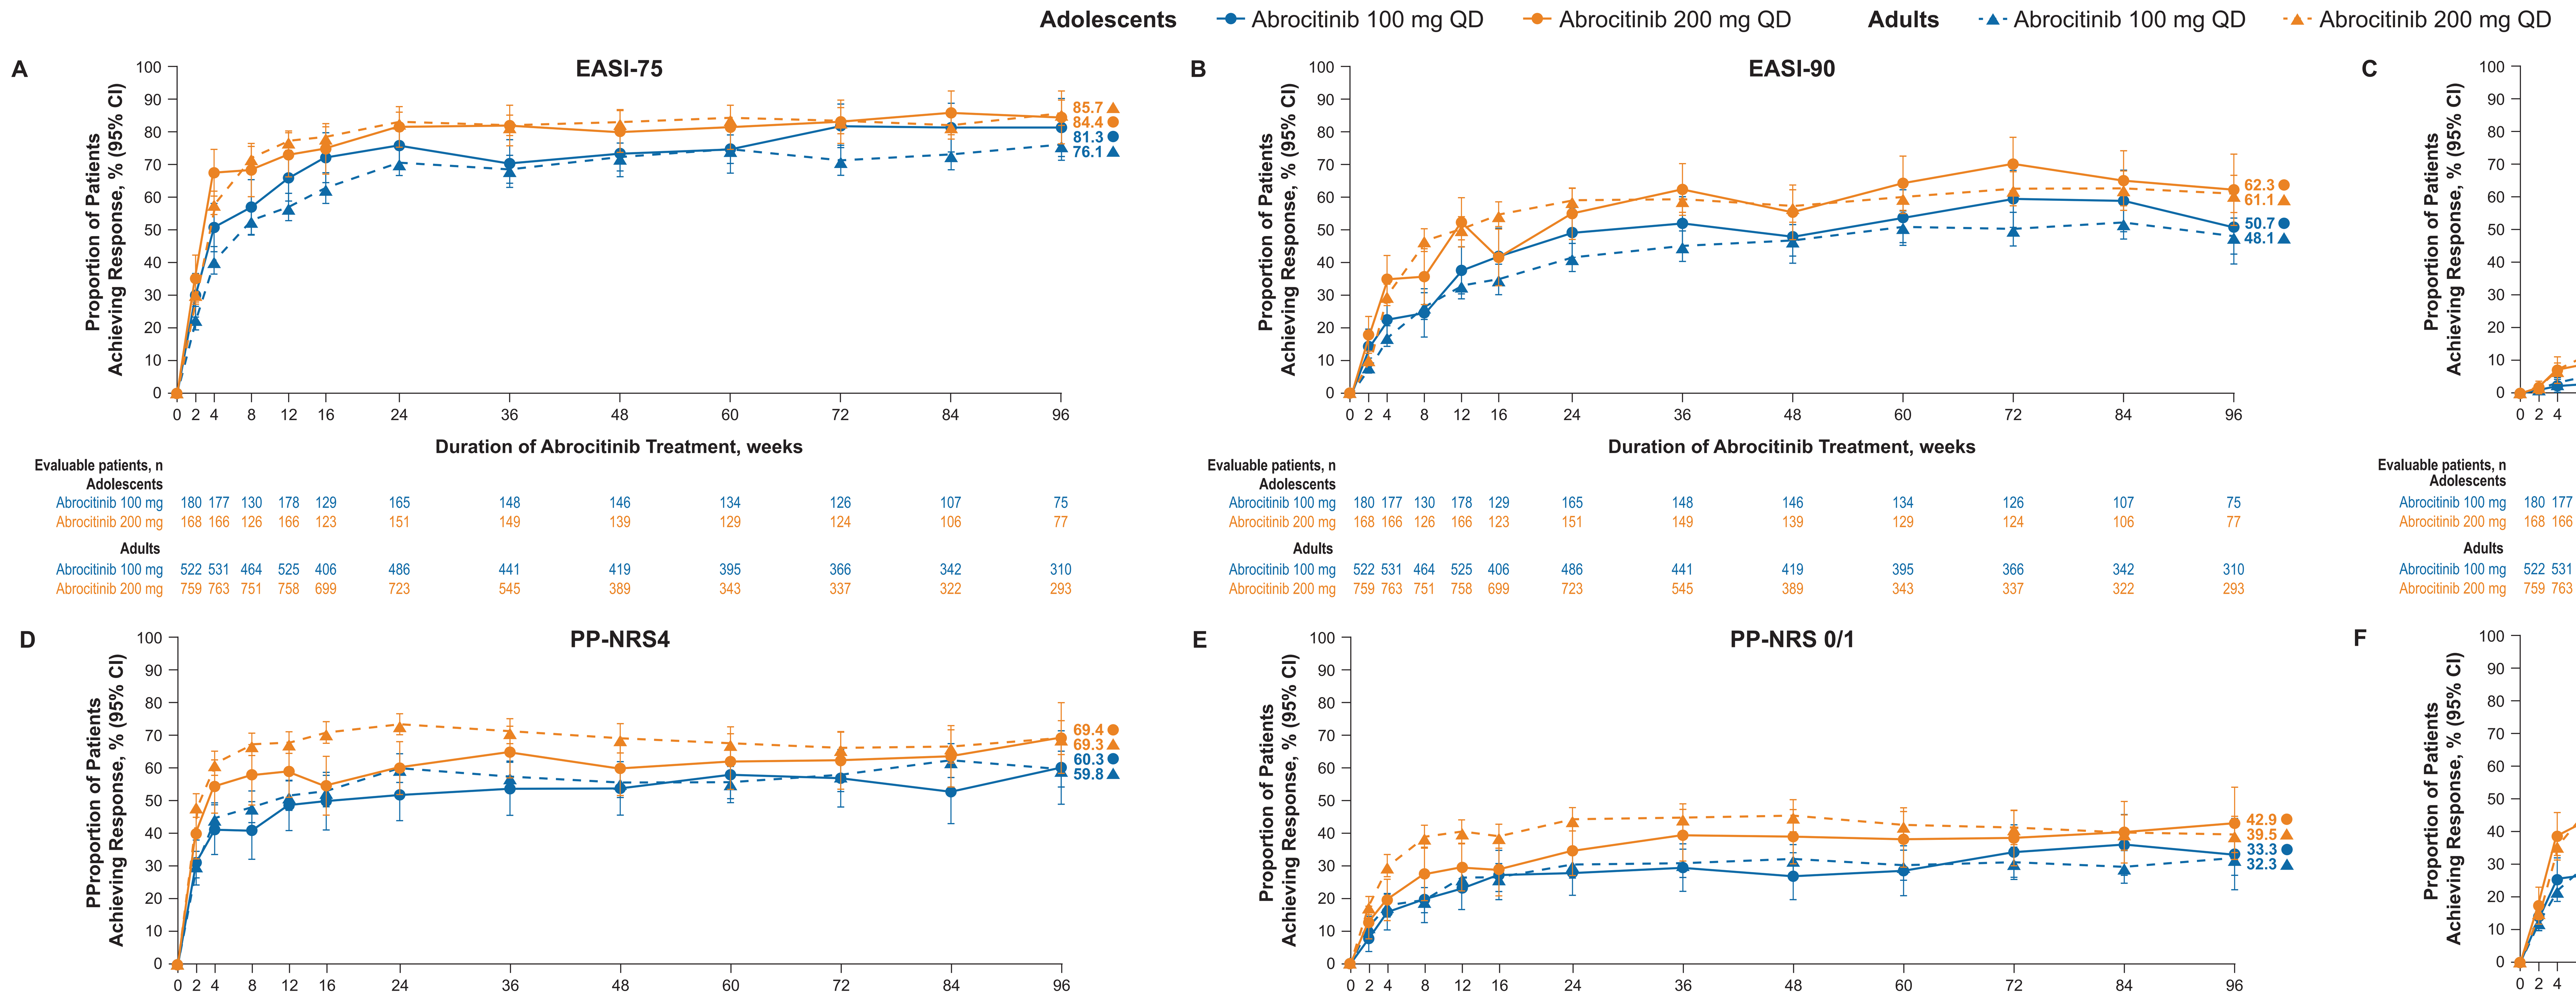
Adolescents
Abrocitinib 100 mg QD
Abrocitinib 200 mg QD
Adults
Abrocitinib 100 mg QD
Abrocitinib 200 mg QD
A
EASI-75
85.7
84.4
81.3
76.1
B
EASI-90
62.3
61.1
50.7
48.1
C
EASI-100
32.5
21.8
18.7
18.7
D
PP-NRS4
69.4
69.3
60.3
59.8
E
PP-NRS 0/1
42.9
39.5
33.3
32.3
F
IGA 0/1
55.9
53.2
49.3
45.1
Proportion of Patients Achieving Response, % (95% CI)
PProportion of Patients Achieving Response, % (95% CI)
Duration of Abrocitinib Treatment, weeks

Evaluable patients, n (A) EASI-75, (B) EASI-90, (C) EASI-100

	2	4	8	12	16	24	36	48	60	72	84	96
Adolescents												
Abrocitinib 100 mg	180	177	130	178	129	165	148	146	134	126	107	75
Abrocitinib 200 mg	168	166	126	166	123	151	149	139	129	124	106	77
Adults												
Abrocitinib 100 mg	522	531	464	525	406	486	441	419	395	366	342	310
Abrocitinib 200 mg	759	763	751	758	699	723	545	389	343	337	322	293

Evaluable patients, n (D) PP-NRS4

	2	4	8	12	16	24	36	48	60	72	84	96
Adolescents												
Abrocitinib 100 mg	176	160	117	164	124	160	145	141	129	121	102	73
Abrocitinib 200 mg	162	145	107	149	117	146	143	135	124	120	102	72
Adults												
Abrocitinib 100 mg	521	468	415	491	401	481	438	411	389	363	341	306
Abrocitinib 200 mg	757	699	711	741	696	729	545	389	344	335	318	290

Evaluable patients, n (E) PP-NRS 0/1

	2	4	8	12	16	24	36	48	60	72	84	96
Adolescents												
Abrocitinib 100 mg	182	165	122	170	129	166	150	146	134	126	107	75
Abrocitinib 200 mg	167	149	110	153	122	151	148	139	129	125	107	77
Adults												
Abrocitinib 100 mg	528	474	420	499	406	489	446	419	395	368	345	310
Abrocitinib 200 mg	763	703	717	747	702	735	551	392	347	339	322	294

Evaluable patients, n (F) IGA 0/1

	2	4	8	12	16	24	36	48	60	72	84	96
Adolescents												
Abrocitinib 100 mg	180	176	129	178	129	165	148	145	134	126	107	75
Abrocitinib 200 mg	167	166	126	166	123	150	148	139	129	123	106	77
Adults												
Abrocitinib 100 mg	523	532	463	525	402	482	436	418	394	364	342	308
Abrocitinib 200 mg	763	763	752	759	699	723	543	387	344	336	322	290

EASI-75, ≥75% improvement from baseline in Eczema Area and Severity Index; EASI-90, ≥90% improvement from baseline in Eczema Area and Severity Index; EASI-100, 100% improvement from baseline in Eczema Area and Severity Index; IGA 0/1, Investigator's Global Assessment score of 0 (clear) or 1 (almost clear) with a ≥2-grade improvement from baseline; PP-NRS4, ≥4-point improvement from baseline in Peak Pruritus Numerical Rating Scale score; PP-NRS 0/1, Peak Pruritus Numerical Rating Scale score of 0 (no itch) or 1 (very little itch); QD, once daily.

Data are reported as observed. Data cut-off date: September 25, 2021 (JADE EXTEND is an ongoing trial, and not all patients reached week 96 visits at the clinical data cutoff). For all patients, the baseline value is always the last pre-dose observation, taken from the qualifying parent trial. Day 1 of treatment was defined as the first dose of abrocitinib received by the patient in either the qualifying parent trial or JADE EXTEND. PP-NRS used with permission from Regeneron Pharmaceuticals, Inc. ,and Sanofi

REFERENCES

1. Cibinqo (abrocitinib). Prescribing information. Pfizer Labs; February 2023.
2. Cibinqo (abrocitinib) 100 mg film-coated tablets. Summary of product characteristics. Pfizer Ltd.; March 10, 2023.
3. Simpson EL et al. *Lancet*. 2020;396:255-266.
4. Silverberg JI et al. *JAMA Dermatol.* 2020;156:863-873.
5. Bieber T et al. *N Engl J Med.* 2021;384:1101-1112.
6. Eichenfield LF et al. *JAMA Dermatol.* 2021;157:1165-1173.

ACKNOWLEDGMENTS

Editorial/medical writing support under the guidance of authors was provided by Megan K. Elder, PhD, at ApotheCom, San Francisco, CA, USA, and was funded by Pfizer Inc., New York, NY, USA, in accordance with Good Publication Practice (GPP 2022) guidelines (*Ann Intern Med.* 2022; 10.7326/M22-1460).

This study was funded by Pfizer Inc.

Previously presented at the 103rd Annual Meeting of the British Association of Dermatologists (BAD); June 27-29, 2023; Liverpool, United Kingdom.

DISCLOSURES

ASP is/has served as a study investigator for AbbVie, AnaptysBio, Eli Lilly, Incyte, Janssen, KrystalBio, Regeneron, and UCB; is/has received honoraria as a consultant for Pfizer Inc., Abbvie, Acrotech, Almirall, Amgen, Amryt, Arcutis, Arena, Azitra, BioCryst, BiomX, Bridgebio, Bristol Myers Squibb, Castle Biosciences, Catawba, Eli Lilly, Exicure, Gilead, Incyte, Janssen, Johnson & Johnson, Kamari, Leo, Novartis, OM Pharma, Pierre Fabre, RAPT, Regeneron, Sanofi/Genzyme, Seanergy, and UCB; and is/has served on the Data Safety Monitoring Board for AbbVie, Abeona, Bausch, Galderma, and Novan. **MJC** has been a clinical trial investigator for Pfizer Inc., Atopix, Galapagos, Hyphens, Johnson & Johnson, Kymab, LEO Pharma, L'Oreal/La Roche-Posay, Novartis, Regeneron, and Sanofi Genzyme and an advisory board member, consultant, and/or invited lecturer for Pfizer Inc., Abbvie, Amlar, Astellas, Atopix, Boots, Dermavant, Galapagos, Galderma, Hyphens, Johnson & Johnson, Kymab, LEO Pharma, L'Oreal/La Roche-Posay, Menlo Therapeutics, Novartis, Oxagen, Procter & Gamble, Reckitt Benckiser, Regeneron, and Sanofi-Genzyme. **CF** is chief investigator of the UK National Institute for Health Research-funded TREAT (ISRCTN15837754) and SOFTER (Clinicaltrials.gov: NCT03270566) trials as well as the UK-Irish Atopic eczema Systemic Therapy Register (A-STAR; ISRCTN11210918) and a principal investigator in the European Union Horizon 2020-funded BIOMAP Consortium (http://www.biomap-imi.eu/). His department has also received funding from Sanofi-Genzyme. **CB** is/has been a clinical trial investigator for Abbvie, Eli Lilly, Galderma, Merck, Novartis, and Sanofi-Genzyme and an advisory board member, consultant and/or invited lecturer for Abbvie, ALK, Eli Lilly, LEO Pharma, Mylan, Merck, Novartis, Pfizer, and Sanofi-Genzyme. **SW** has received institutional research grants from Sanofi Deutschland GmbH, Leo Pharma, and La Roche Posay; has performed consultancies for Sanofi-Genzyme, Regeneron, LEO Pharma, AbbVie, Pfizer, Eli Lilly, Kymab, and Novartis; has lectured at educational events sponsored by Sanofi Genzyme, Regeneron, LEO Pharma, AbbVie, Novartis, and Galderma; and is involved in performing clinical trials with many pharmaceutical industries that manufacture drugs used for the treatment of psoriasis and atopic dermatitis. **JN**, **SAF**, **PB**, **MW** and **HK** are employees and shareholders of Pfizer Inc.

Copies of this poster and supplemental information obtained through this QR code are for your personal use only and may not be reproduced without permission from the authors.

Copyright © 2023. All rights reserved.

Presented at the Fall Clinical Dermatology Conference; October 19-22, 2023; Las Vegas, Nevada